Costimulatory Antigen Receptor (CoStAR): A Novel Platform That Enhances the Activity of Tumor-Infiltrating Lymphocytes

Poster **198**

Sujita Sukumaran,[1] Milena Kalaitsidou,[1] Michelle Mojadidi,[1] Clare Yarka,[1] Yong (Stella) Ouyang,[1] Eric Gschweng,[1] Gray Kueberuwa,[1] John S. Bridgeman,[1] Robert E. Hawkins,[1] and Rubén Alvarez-Rodríguez[1]

[1]*Instil Bio, Inc, Dallas, TX, USA*

InstilBio

BACKGROUND

- Major limiting factors for development of cell therapy modalities in solid tumors include clonal heterogeneity and the related lack of universally expressed tumor-specific antigens[1-3]
- As the only truly polyclonal cell product in advanced development, tumor-infiltrating lymphocytes (TILs) encompass the maximum possible diversity available with an unrestricted T-cell receptor repertoire (TCR), thereby offering the broadest diversity of anti-tumor reactivity[4,5]
- The suppressive tumor microenvironment is characterized by high expression of coinhibitory receptors and poor costimulation[6,7]
- To overcome poor endogenous costimulation, a novel platform was engineered to leverage the diverse TCR repertoire of TILs while amplifying their antitumor activity via a synthetic costimulatory antigen receptor (CoStAR; **Figure 1**)

Figure 1. CoStAR Platform Overview

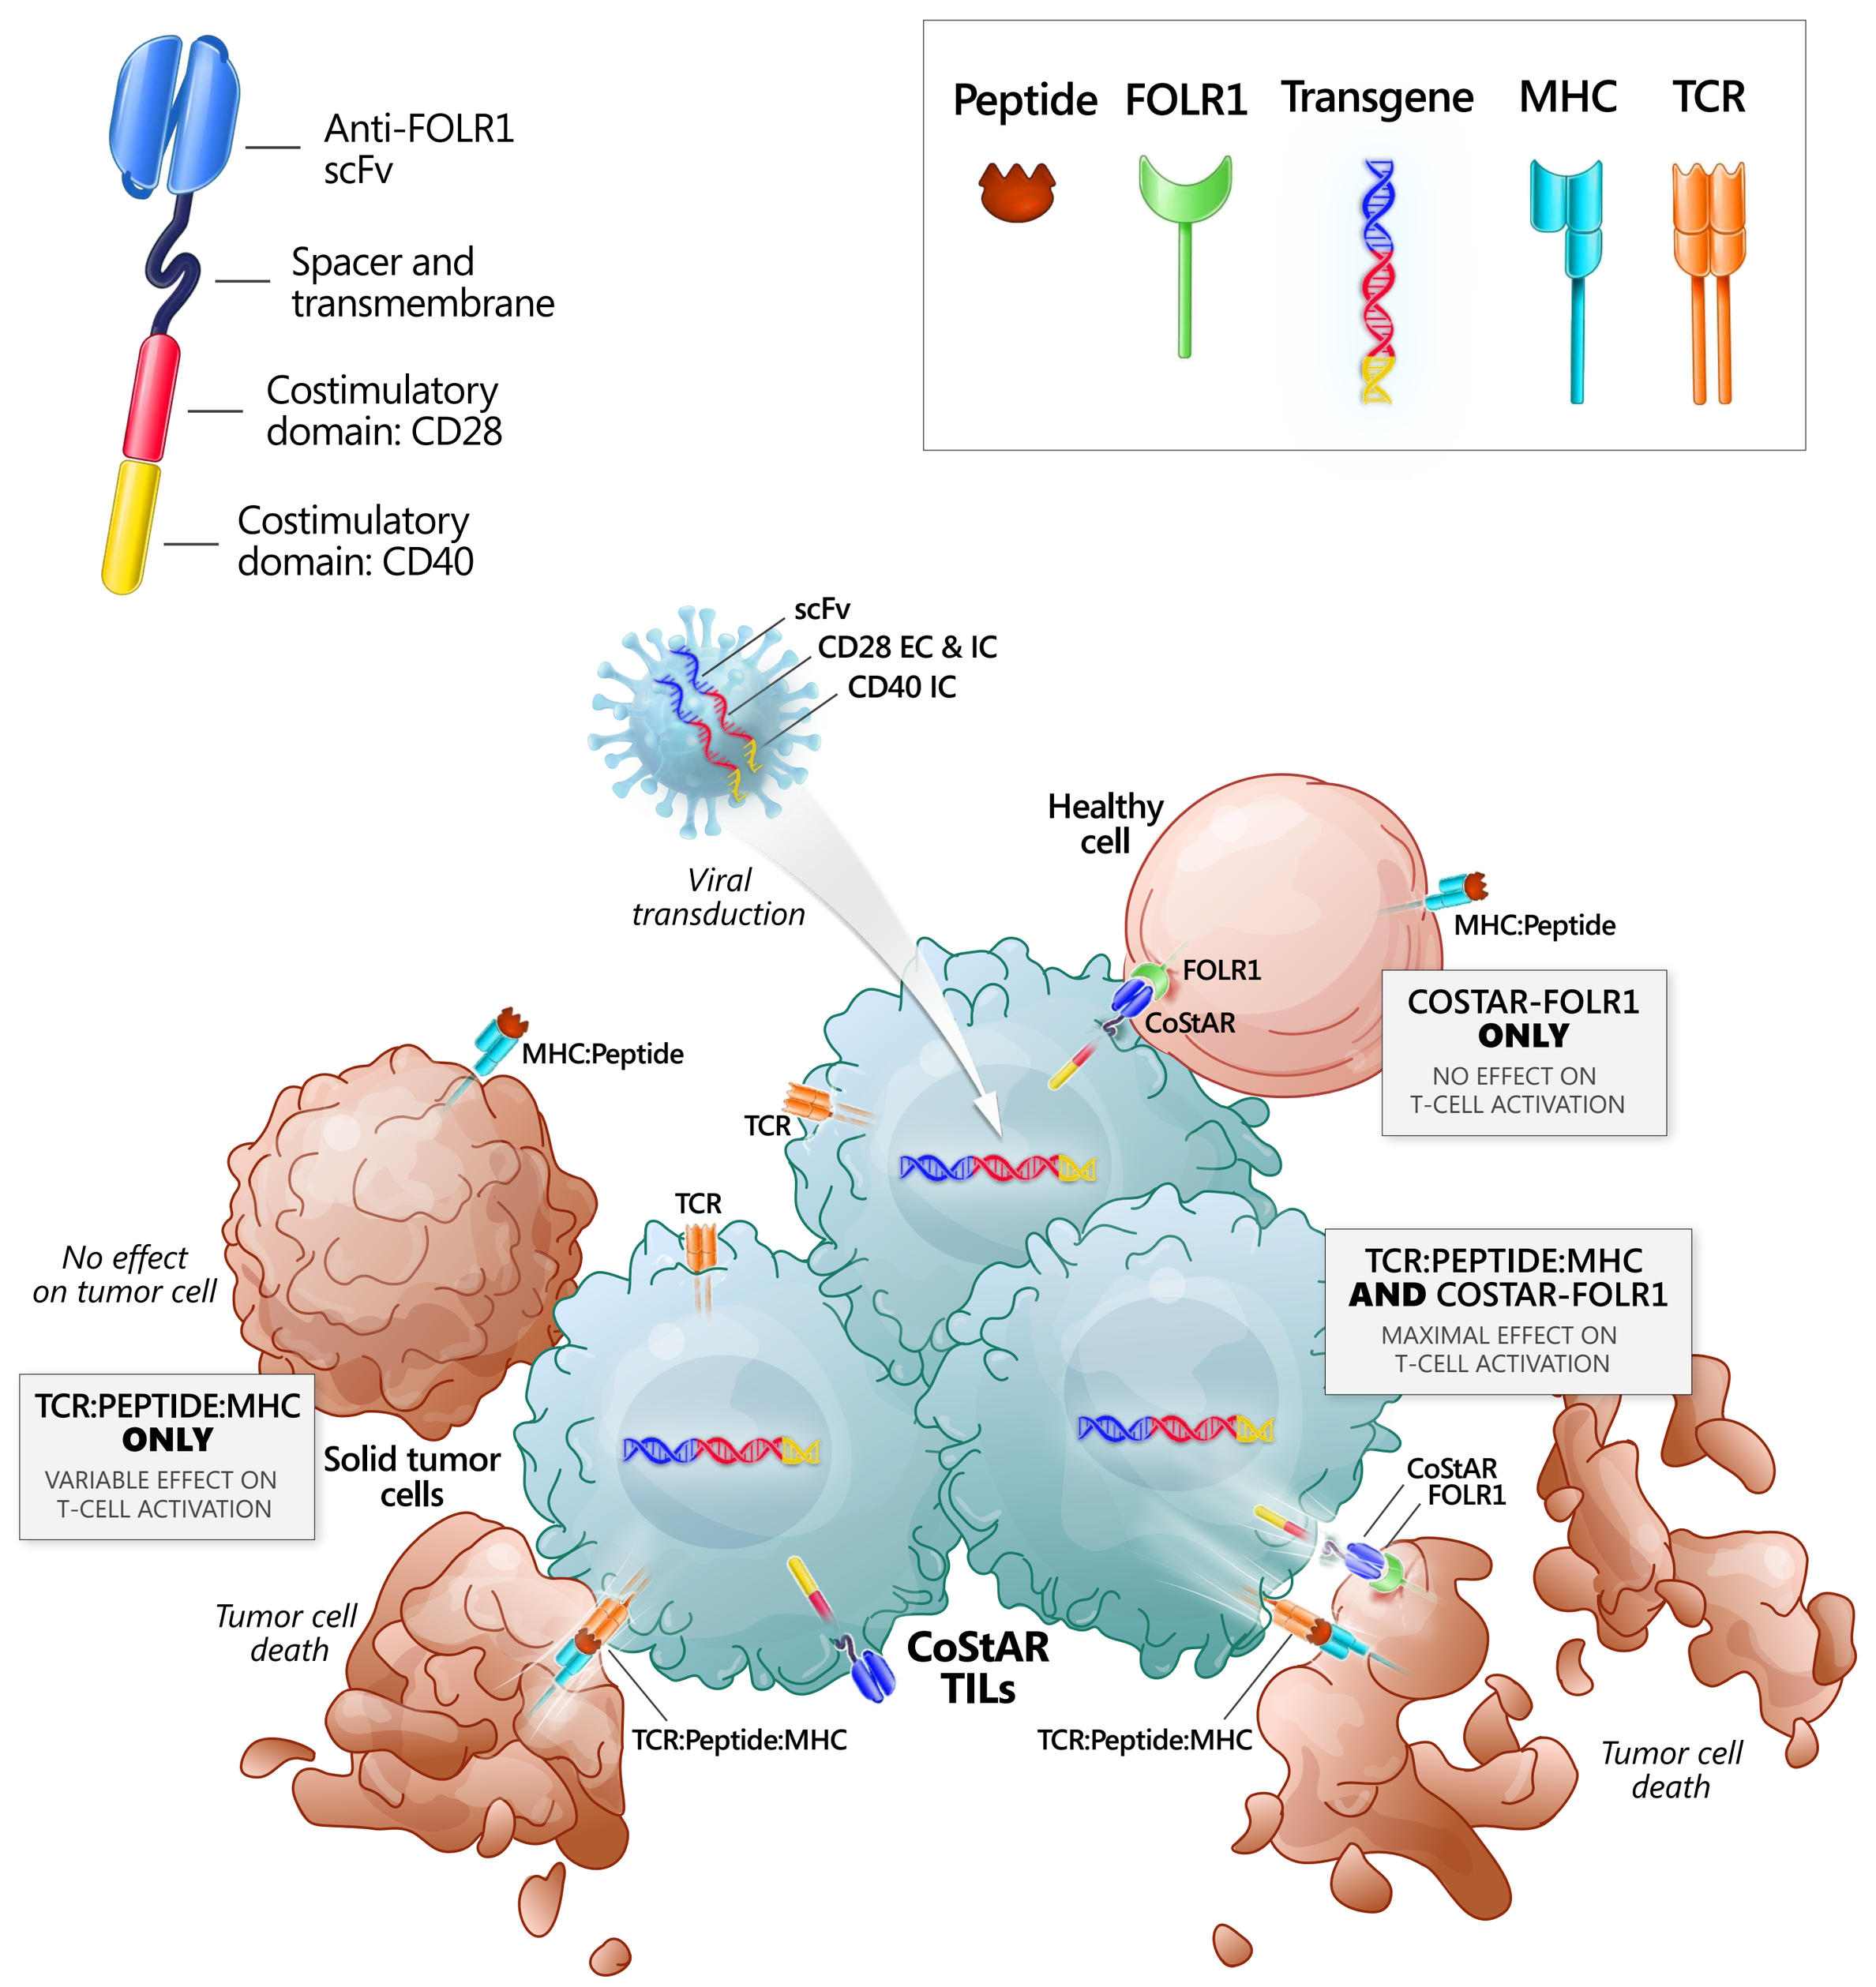
Anti-FOLR1 scFv
Spacer and transmembrane
Costimulatory domain: CD28
Costimulatory domain: CD40
Peptide FOLR1 Transgene MHC TCR
scFv
CD28 EC & IC
CD40 IC
Viral transduction
Healthy cell
MHC:Peptide
FOLR1
CoStAR
COSTAR-FOLR1 ONLY
NO EFFECT ON T-CELL ACTIVATION
TCR
No effect on tumor cell
TCR:PEPTIDE:MHC ONLY
VARIABLE EFFECT ON T-CELL ACTIVATION
Solid tumor cells
TCR:PEPTIDE:MHC AND COSTAR-FOLR1
MAXIMAL EFFECT ON T-CELL ACTIVATION
Tumor cell death
CoStAR TILs
TCR:Peptide:MHC

CoStAR, costimulatory antigen receptor; EC, extracellular; FOLR1, folate receptor alpha; IC, intracellular; MHC, major histocompatibility complex; scFv, single-chain variable fragment; TCR, T-cell receptor; TIL, tumor-infiltrating lymphocyte.

METHODS

- A CoStAR molecule encoding an extracellular folate receptor alpha (FOLR1)-targeting single-chain fragment variable (scFv) and intracellular CD28 and CD40 signaling sequences was transduced with a lentiviral vector into peripherally harvested T cells from 3 healthy donors and TILs derived from 5 primary ovarian tumors
- Anti-FOLR1 CoStAR-expressing T cells were cocultured with target cells expressing:
 - Membrane-anchored anti-CD3 antibody (OKT3) to provide Signal 1 through TCR/CD3 complex crosslinking
 - FOLR1 to provide potent costimulatory Signal 2 through CoStAR
- T-cell activity was measured by quantifying expression of activation markers, cytokine secretion, T-cell proliferation, and cytolytic activity compared with a non-transduced control

RESULTS

Figure 2. FOLR1 Protein Expression Profile in Selected Tumors

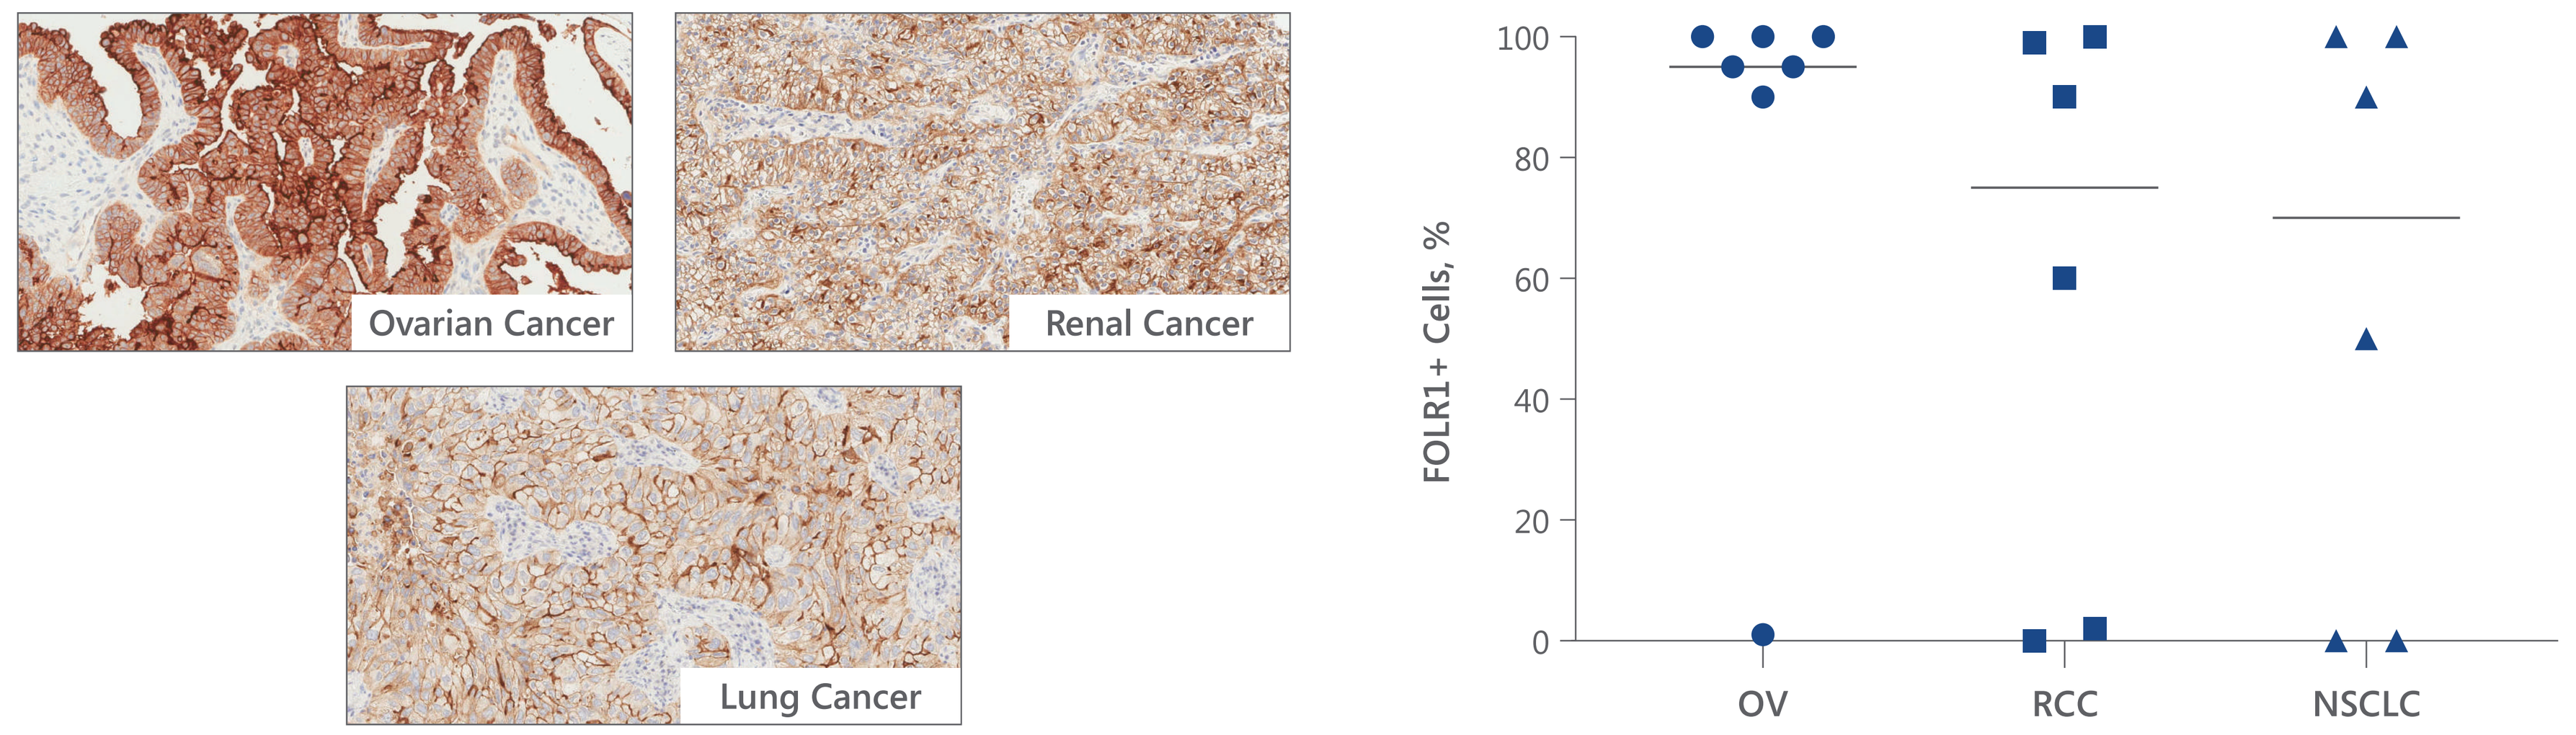
Ovarian Cancer
Renal Cancer
Lung Cancer
FOLR1+ Cells, %
OV
RCC
NSCLC

FOLR1, folate receptor alpha; NSCLC, non-small cell lung cancer (adenocarcinoma); OV, ovarian cancer (high-grade serous); RCC, renal cell carcinoma (clear cell).

- Robust protein expression of FOLR1 was observed in ovarian, renal, and lung cancer histologies

Figure 3. Ovarian, Renal, and Lung TILs Demonstrated Reactivity Against Autologous Tumor

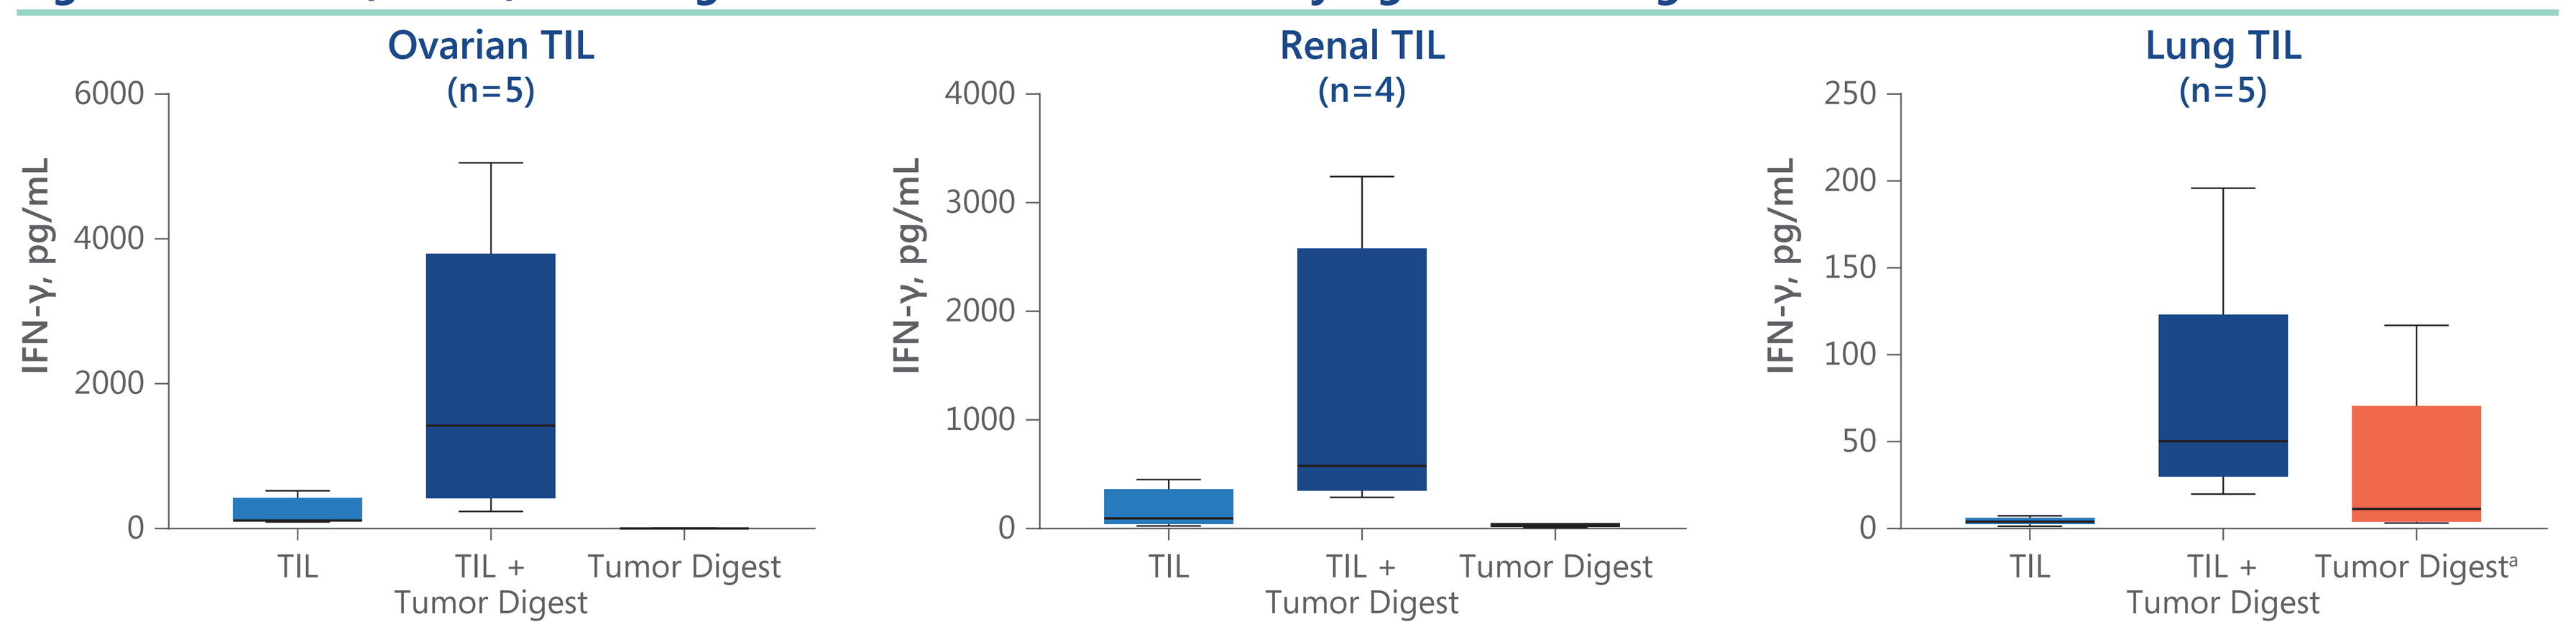
Ovarian TIL (n=5)
Renal TIL (n=4)
Lung TIL (n=5)
IFN-γ, pg/mL
TIL
TIL + Tumor Digest
Tumor Digest
Tumor Digest[a]

[a] Lung tumor digests contain more T cells compared with other histologies, which results in background.
IFN, interferon; TIL, tumor-infiltrating lymphocyte.

- TILs isolated and manufactured from different tumor types showed interferon (IFN)-γ production when cocultured with ovarian, renal, and lung autologous tumor digests
- Comprehensive analysis in human T cells from 3 healthy donors and TILs isolated from 5 ovarian tumors are presented hereafter

Figure 4. Marked Enhancement of Effector Function in Anti-FOLR1 CoStAR-Expressing T Cells

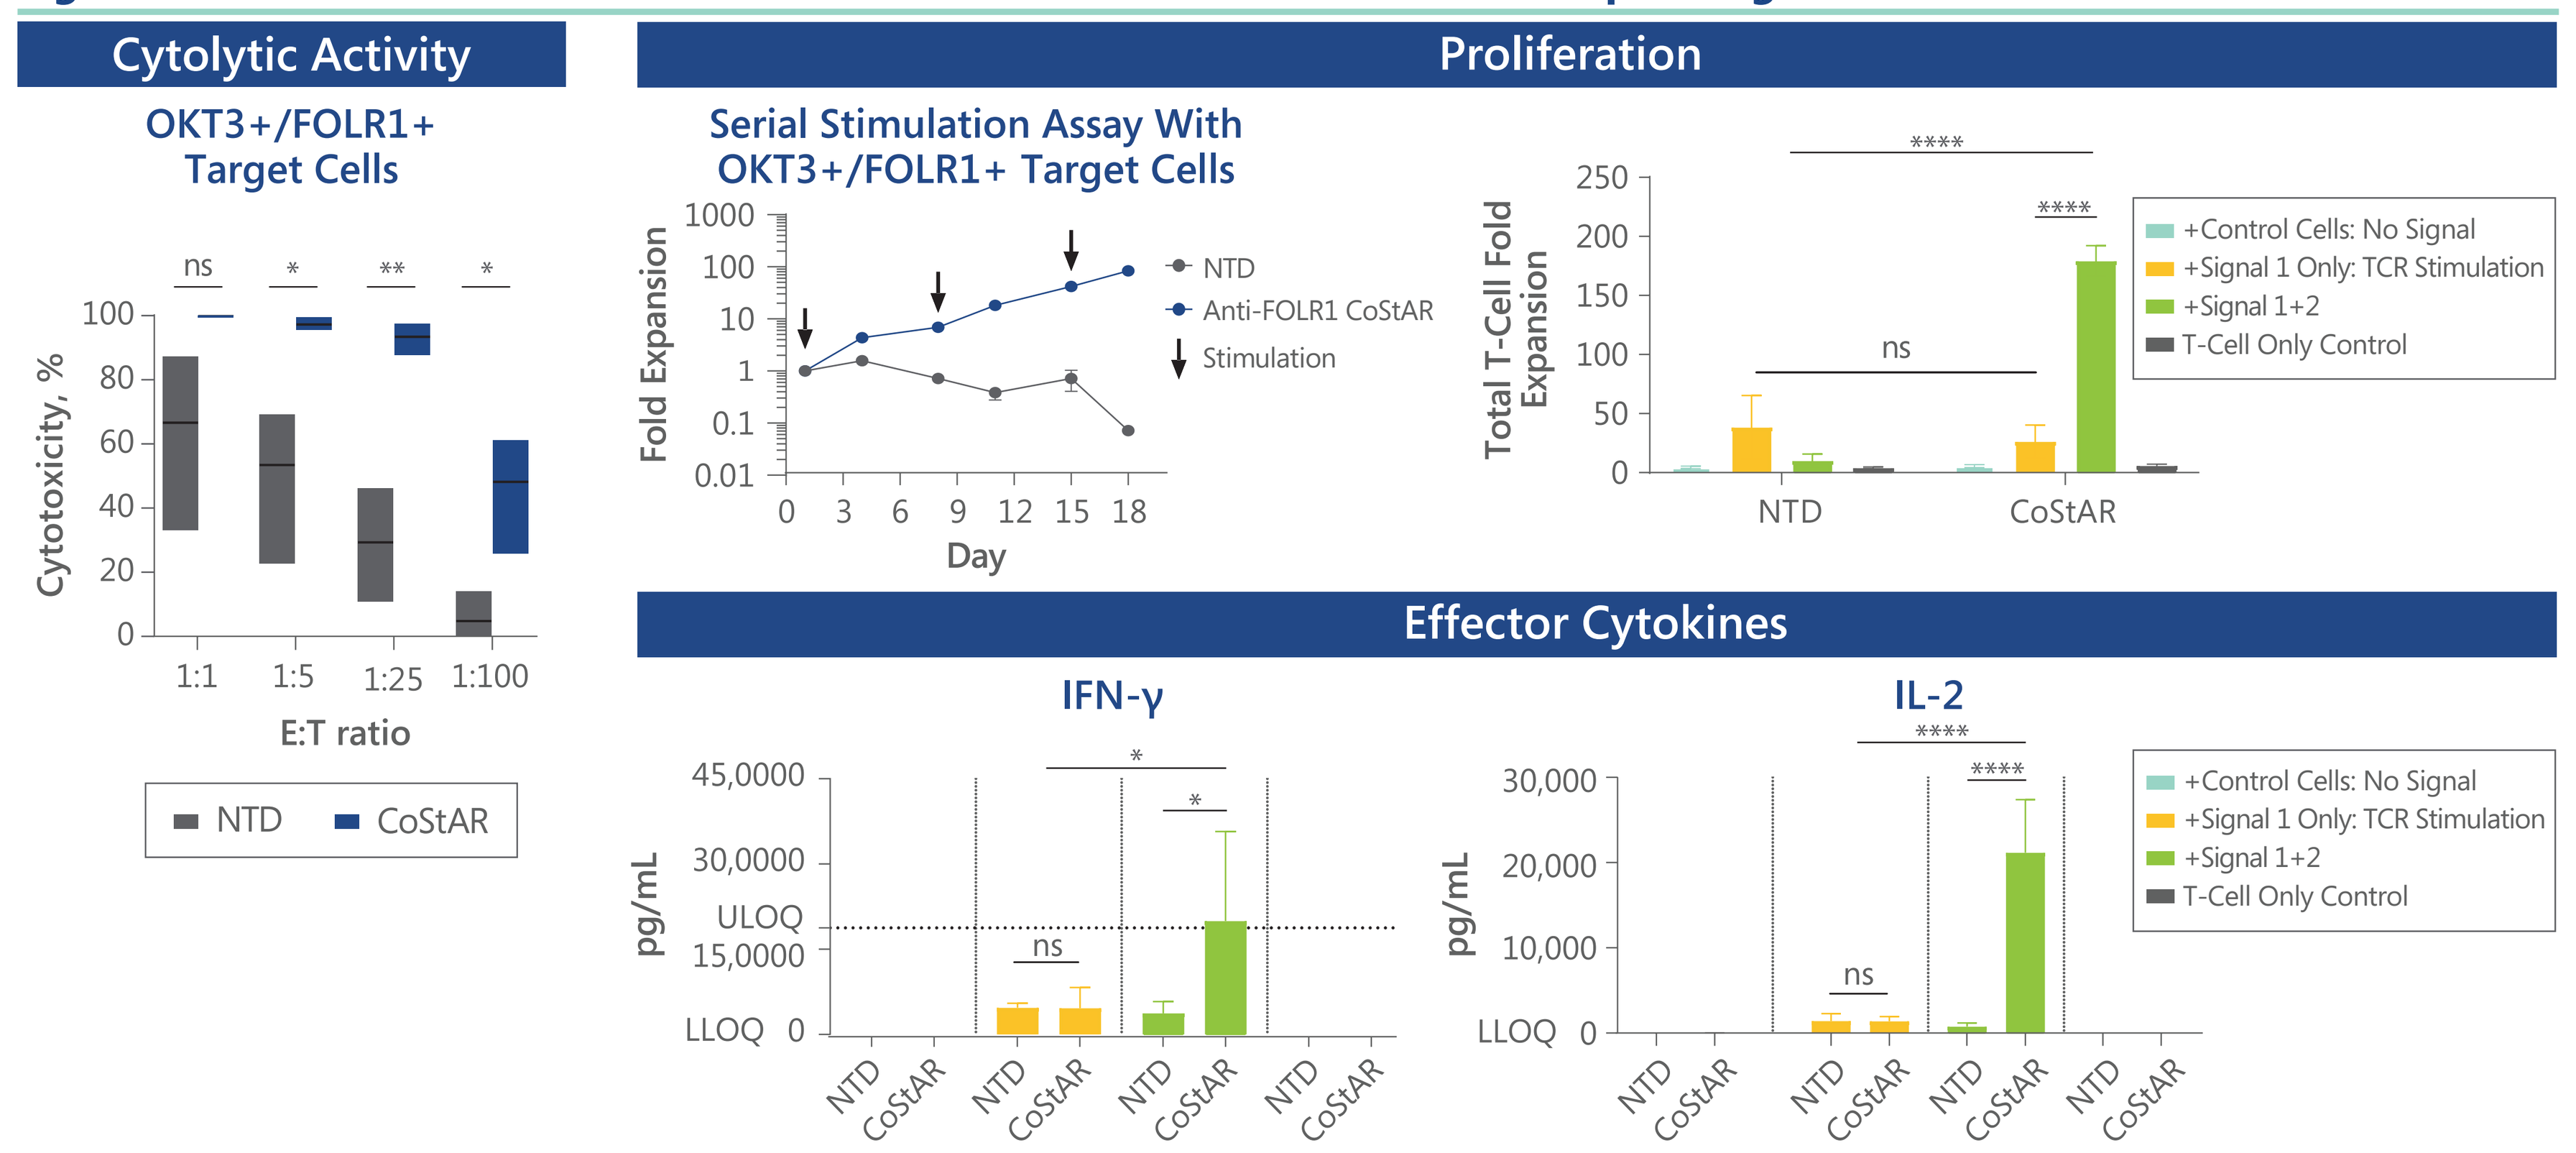
Cytolytic Activity
OKT3+/FOLR1+ Target Cells
Cytotoxicity, %
E:T ratio
NTD
CoStAR
Proliferation
Serial Stimulation Assay With OKT3+/FOLR1+ Target Cells
Fold Expansion
Day
NTD
Anti-FOLR1 CoStAR
Stimulation
Total T-Cell Fold Expansion
+Control Cells: No Signal
+Signal 1 Only: TCR Stimulation
+Signal 1+2
T-Cell Only Control
Effector Cytokines
IFN-γ
IL-2
pg/mL
ULOQ
LLOQ

Statistical significance was assessed by 2-way analysis of variance test; ns, $P>.1$, *$P<.0275$, **$P<.005$, and ****$P<.0001$.
CoStAR, costimulatory antigen receptor; E:T, effector:target; FOLR1, folate receptor alpha; IFN, interferon; IL, interleukin; LLOQ, lower limit of quantitation; ns, not significant; NTD, non-transduced; TCR, T-cell receptor; ULOQ, upper limit of quantitation.

- Healthy donor T cells were transduced with anti-FOLR1 CoStAR and cocultured with target cells expressing membrane-bound OKT3 with or without FOLR1
- CoStAR-expressing T cells performed equivalently to non-transduced T cells when cocultured with target cells that did not express FOLR1
- A dramatic improvement was seen in cytolytic activity, proliferation, and cytokine secretion by anti-FOLR1 CoStAR-expressing T cells compared with non-transduced T cells

Figure 5. Binding of FOLR1 by CoStAR Alone Does Not Trigger Activation

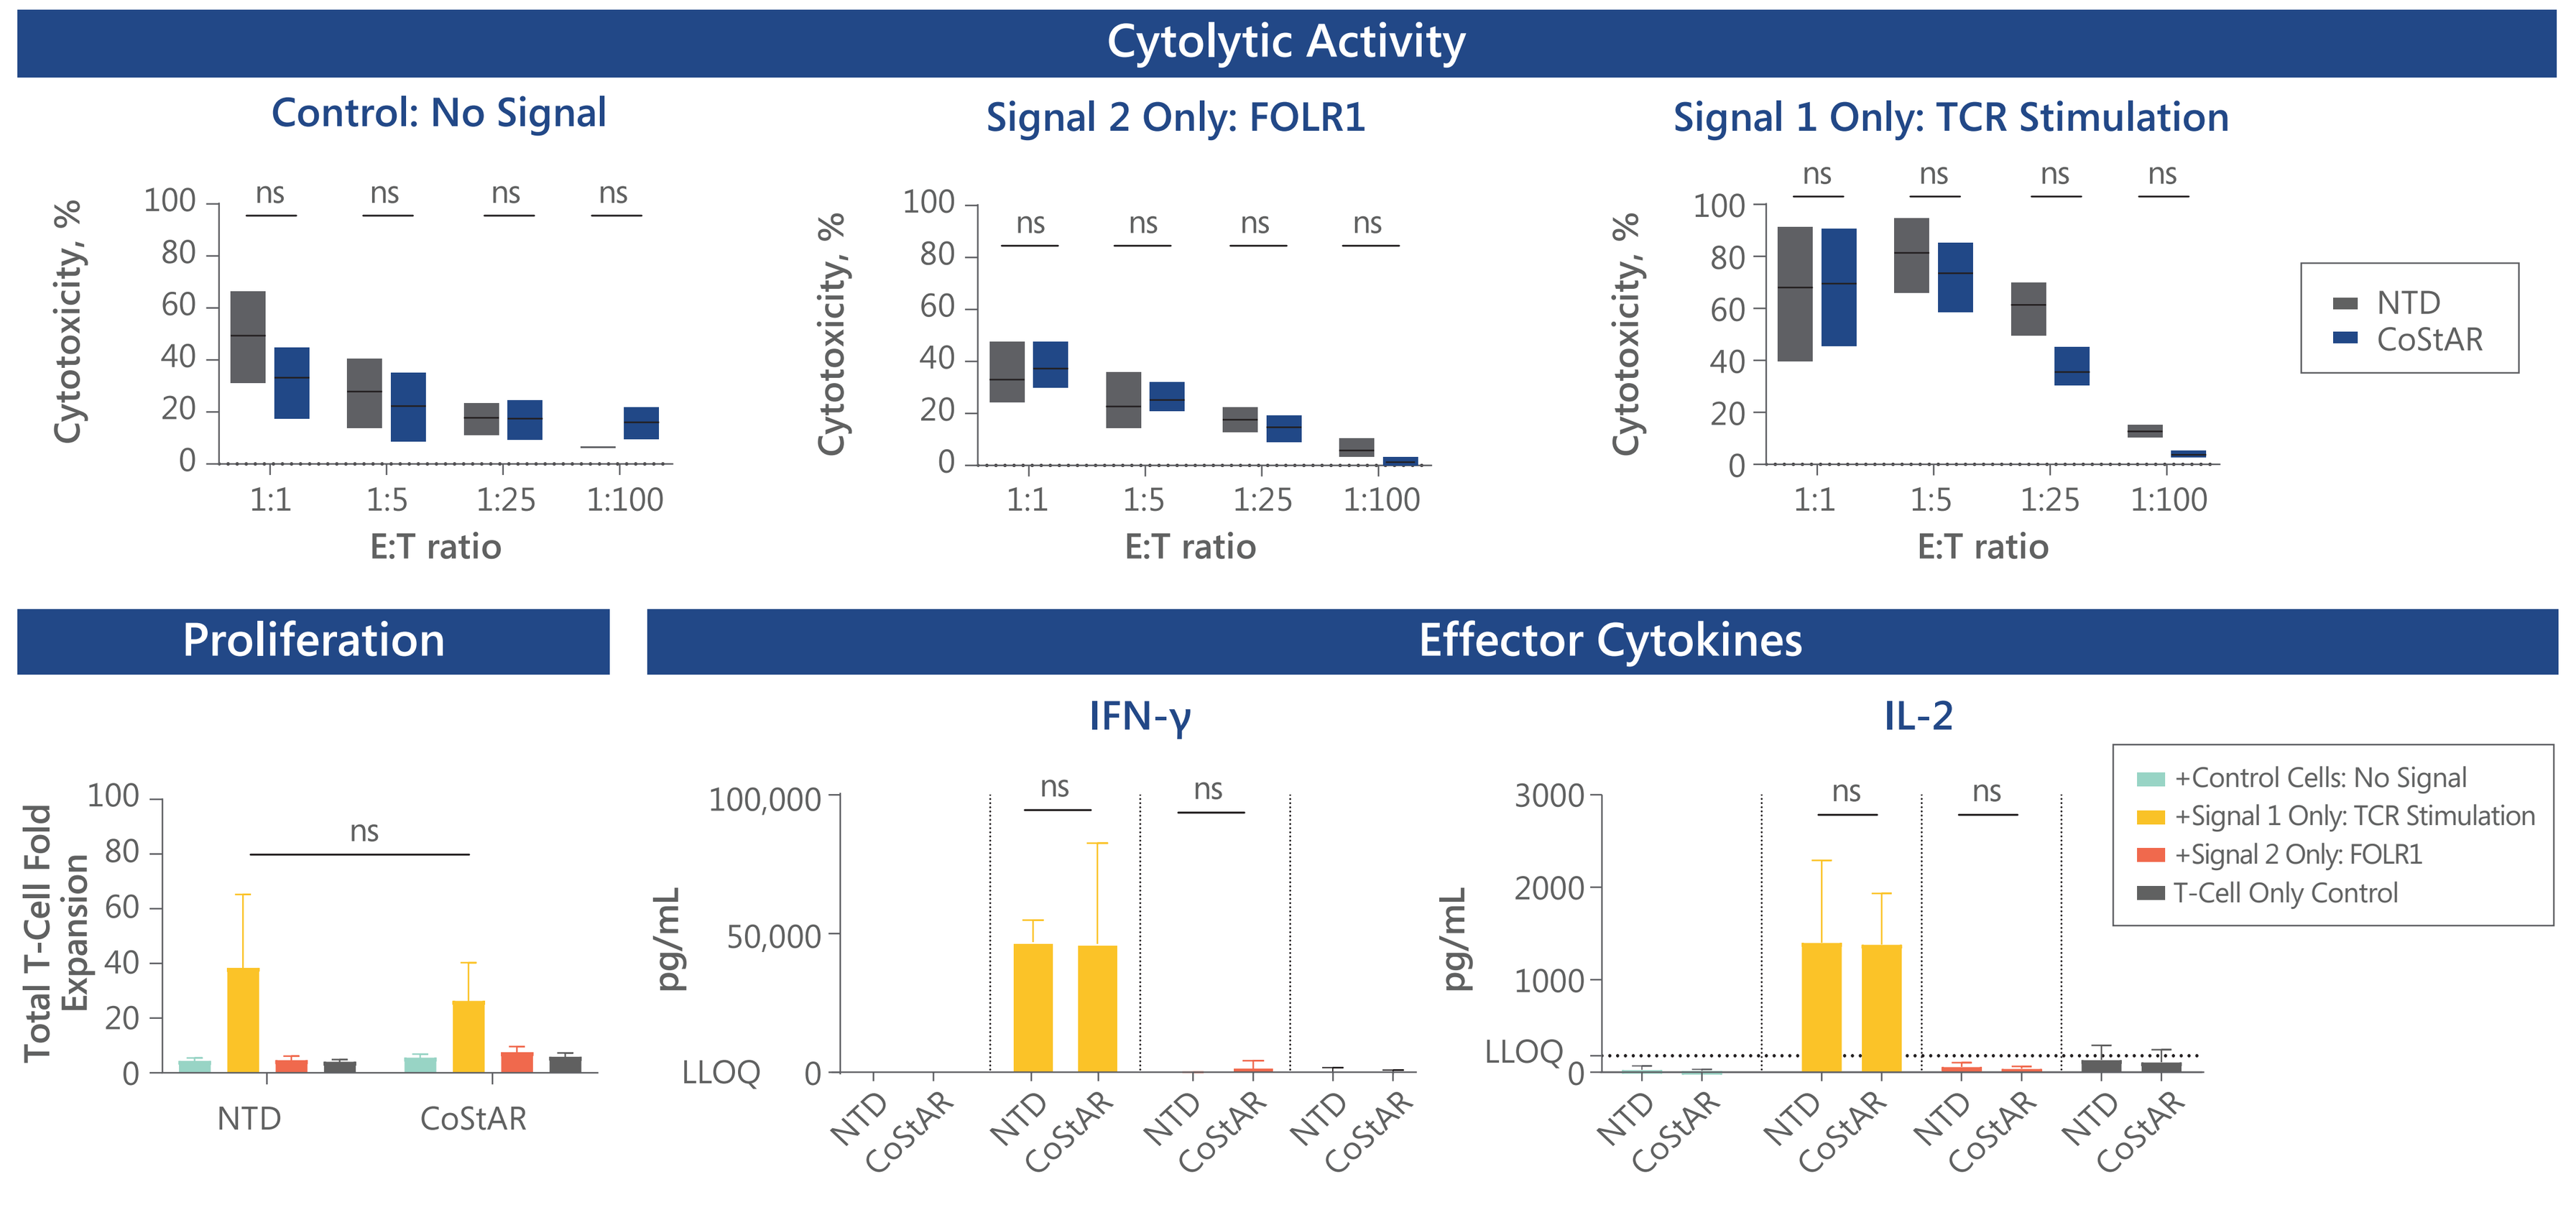
Cytolytic Activity
Control: No Signal
Signal 2 Only: FOLR1
Signal 1 Only: TCR Stimulation
Cytotoxicity, %
E:T ratio
NTD
CoStAR
Proliferation
Total T-Cell Fold Expansion
Effector Cytokines
IFN-γ
IL-2
pg/mL
LLOQ
+Control Cells: No Signal
+Signal 1 Only: TCR Stimulation
+Signal 2 Only: FOLR1
T-Cell Only Control

Statistical significance was assessed by 2-way analysis of variance test; ns, P>.9.
CoStAR, costimulatory antigen receptor; E:T, effector:target; FOLR1, folate receptor alpha; IFN, interferon; IL, interleukin; LLOQ, lower limit of quantitation; ns, not significant; NTD, non-transduced; TCR, T-cell receptor.

- Healthy donor T cells were transduced with anti-FOLR1 CoStAR and cocultured with target cells expressing membrane bound FOLR1 or OKT3 alone
- Anti-FOLR1 CoStAR-expressing T cells demonstrated no increased effector activity over non-transduced T cells when exposed to FOLR1+/OKT3- target cells
- Anti-FOLR1 CoStAR-expressing T cells retained equivalent effector function compared with non-transduced T cells when exposed to FOLR1-/OKT3+ target cells

Figure 6. CoStAR Transduction Does Not Alter TIL Differentiation Status

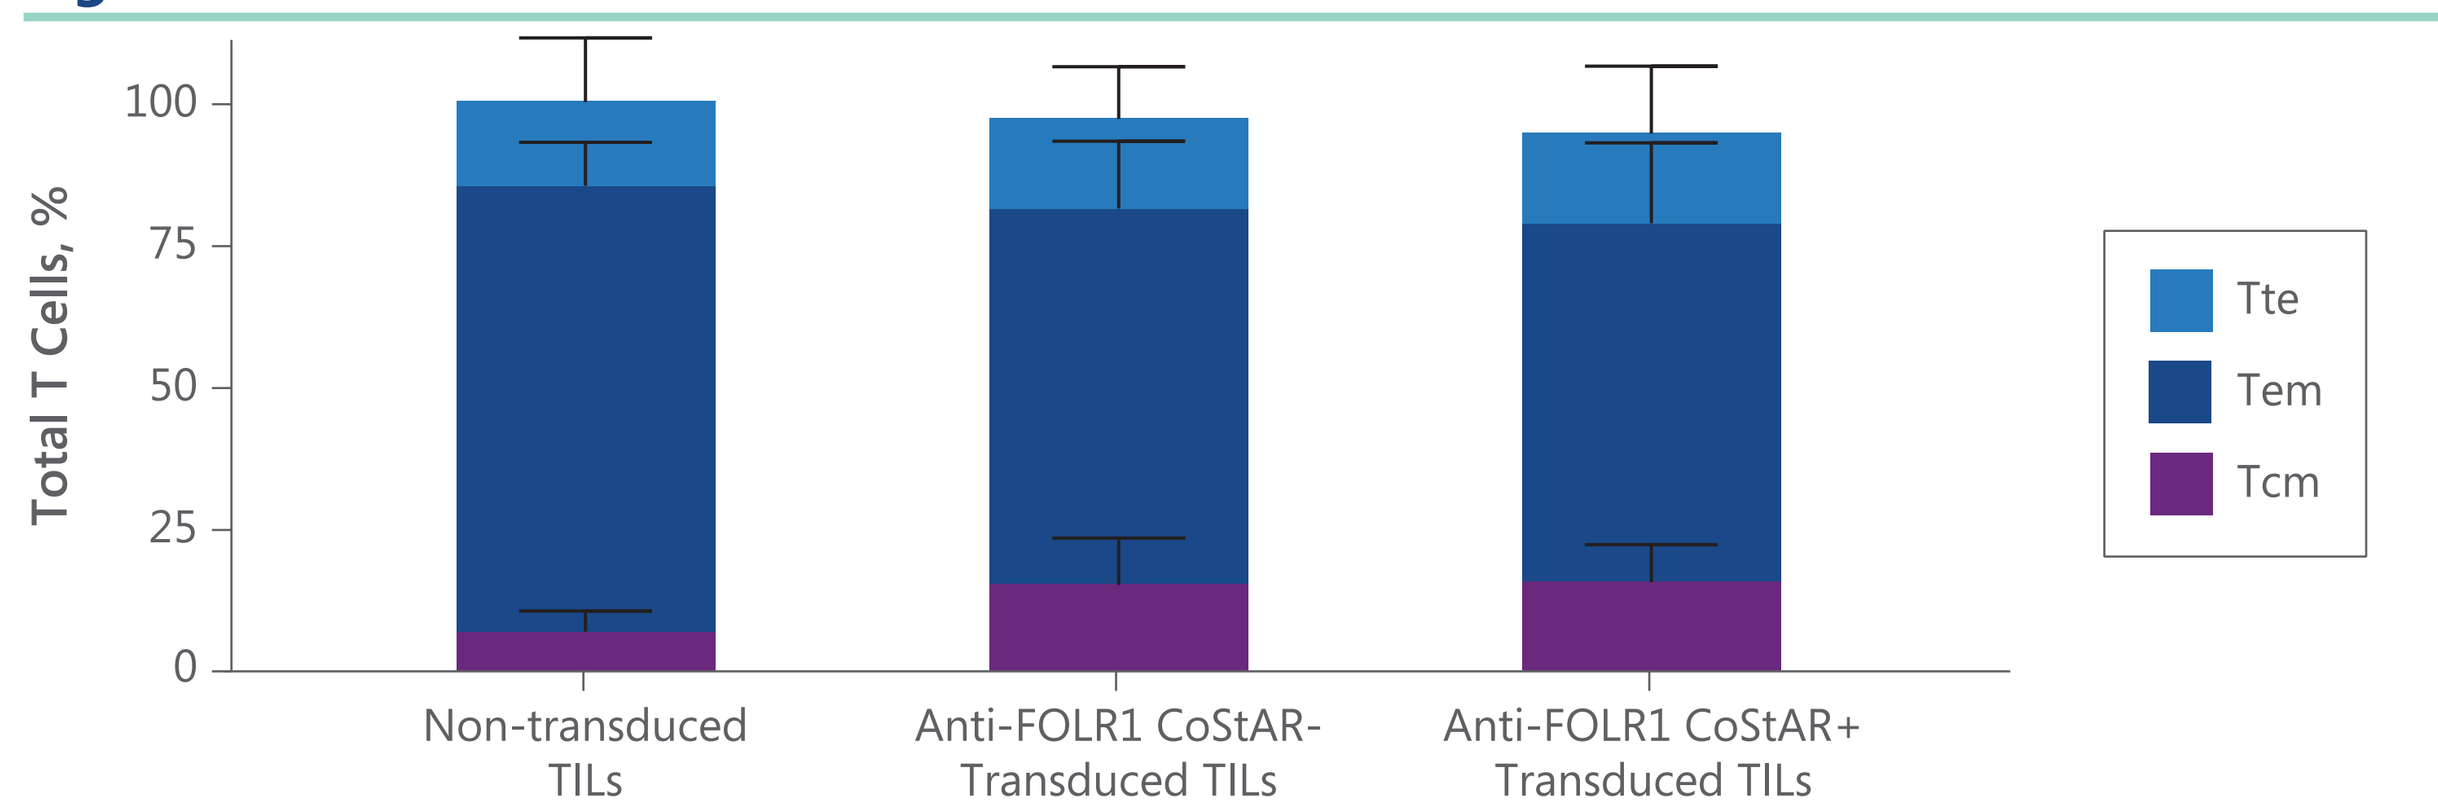
Total T Cells, %
Non-transduced TILs
Anti-FOLR1 CoStAR- Transduced TILs
Anti-FOLR1 CoStAR+ Transduced TILs
Tte
Tem
Tcm

- The phenotype of ovarian TILs, as characterized by differentiation status, was not affected by anti-FOLR1 CoStAR modification

CCR, C-C chemokine receptor; CoStAR, costimulatory antigen receptor; FOLR1, folate receptor alpha; Tcm, central memory T cells (CD45RO+CCR7+CD95+CD27+); Tem, effector memory T cells (CD45RO+CCR7–CD95+CD27+/-); TIL, tumor-infiltrating lymphocyte; Tte, effector T cells (CD45RA+CCR7–CD27–CD95+).

Figure 7. Primary Ovarian TIL Function Is Enhanced by CoStAR Transduction

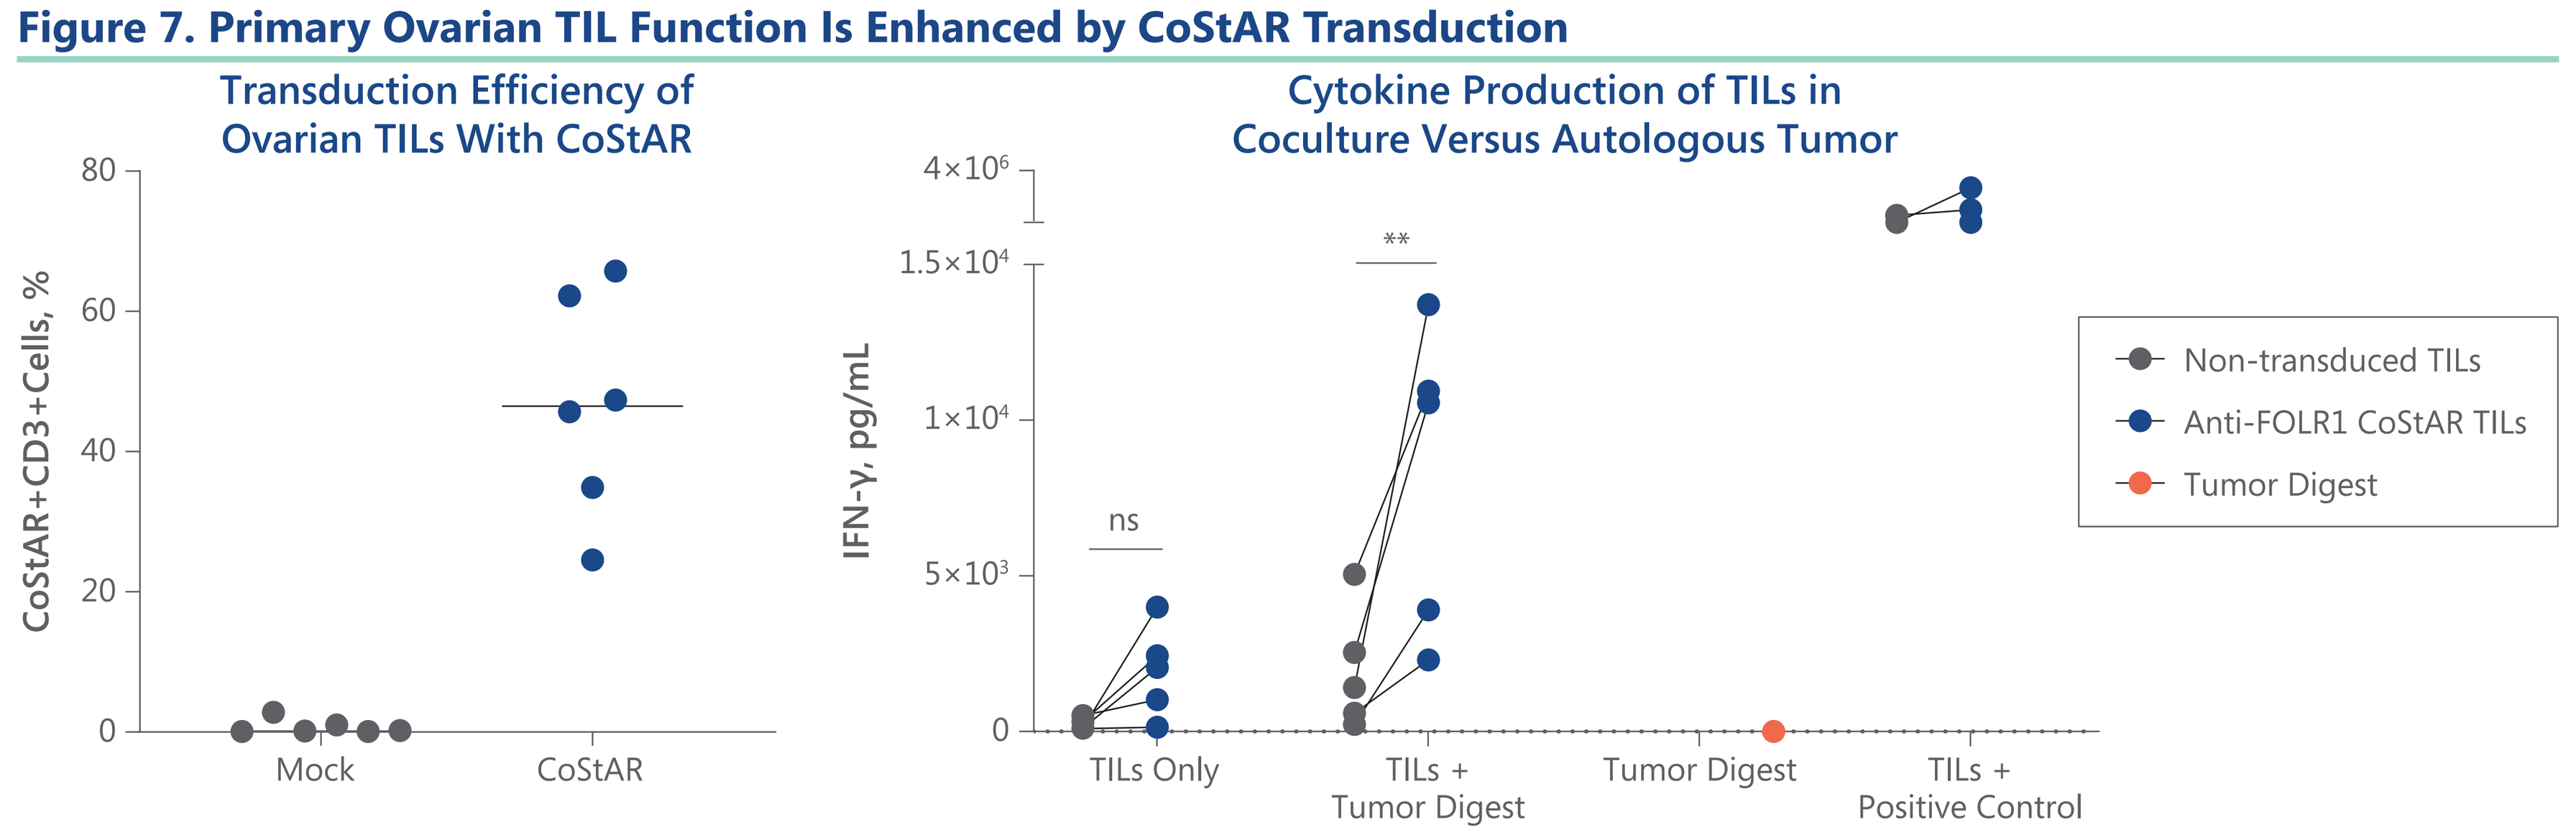
Transduction Efficiency of Ovarian TILs With CoStAR
CoStAR+CD3+Cells, %
Mock
CoStAR
Cytokine Production of TILs in Coculture Versus Autologous Tumor
IFN-γ, pg/mL
TILs Only
TILs + Tumor Digest
Tumor Digest
TILs + Positive Control
Non-transduced TILs
Anti-FOLR1 CoStAR TILs
Tumor Digest

Statistical significance was assessed by 2-way analysis of variance test; ns, $P>.2$ and **$P<.005$.
Phorbol 12-myristate 13-acetate (PMA) and ionomycin were used as a positive control.
CoStAR, costimulatory antigen receptor; FOLR1, folate receptor alpha; IFN, interferon; ns, not significant; TIL, tumor-infiltrating lymphocyte.

- Primary ovarian TILs were efficiently transduced with anti-FOLR1 CoStARs
- Improved cytokine secretion was observed when primary ovarian anti-FOLR1 CoStAR TILs were cocultured with autologous tumor digest

CONCLUSIONS

- The novel costimulatory antigen receptor anti-FOLR1 CoStAR molecule improves T-cell effector function through targeted costimulation upon engagement of tumor-expressed FOLR1
- When combined with concomitant TCR-specific binding, CoStAR's dual intracellular CD28 and CD40 signaling motifs significantly augment cytolytic activity, T-cell proliferation, and cytokine secretion
- Signaling through CoStAR alone does not trigger T-cell effector function
- Anti-FOLR1 CoStAR-expressing T cells retain normal function in the absence of FOLR1 expression on target cells
- Primary human ovarian TIL function is improved by CoStAR
 - For additional information on the CoStAR platform, please see poster 199 by Sykorova et al, titled "Potent T-Cell Costimulation Mediated by a Novel Costimulatory Antigen Receptor With Dual CD28/CD40 Signaling Domains to Improve Adoptive Cell Therapies"
- A first-in-human clinical study with ITIL-306, an investigational anti-FOLR1 CoStAR TIL product, is planned (sponsored by Instil Bio, Inc)
- Additional scFv targets are being evaluated for clinical application across a broad range of solid tumor histologies

REFERENCES

1. Marofi F, et al. *Stem Cell Res Ther*. 2021;12:81.
2. Albelda SM. *Cancer Immunol Res*. 2020;8:2.
3. Shilyansky J, et al. *Proc Natl Acad Sci U S A*. 1994;91:2829-2833.
4. Spindler MJ, et al. *Nat Biotechnol*. 2020;38:609-619.
5. Rohaan MW, et al. *J Immunother Cancer*. 2018;6:102.
6. Truxova I, et al. *J Immunother Cancer*. 2018;6:139.
7. Bandola-Simon J, Roche PA. *Mol Immunol*. 2019;113:31-37.

ACKNOWLEDGMENTS

- Tissue samples were provided by the Cooperative Human Tissue Network, a National Cancer Institute-supported resource. Other investigators may have received samples from these same tissue specimens
- Medical writing support was provided by Christopher Waldapfel, PharmD, of Instil Bio, Inc, and Ashley Skorusa, PhD, and Jennifer Yang, PhD, of Nexus Global Group Science, with funding from Instil Bio, Inc

DISCLOSURES

SS, MK, MM, Y(S)O, GK: employment with and stock or other ownership in Instil Bio, Inc. **CY, EG, RA-R:** employment with and stock or other ownership in Instil Bio, Inc; and pending patent titled, "Methods of Isolating Tumor-Infiltrating Lymphocytes and Uses Thereof." **JSB:** employment with and stock or other ownership in Cellular Therapeutics, Immetacyte, Inc, and Instil Bio, Inc. **REH:** employment with and stock or other ownership in Instil Bio, Inc; and consultancy or advisory role for Anaveon AG, Novalgene, Ltd, Servier, Celyad, Celgene, Oxford Biomedica, GSK, Bristol Myers Squibb, Gilead, EUSA Pharma, Novartis, and Pfizer.

Copies of this poster obtained through Quick Response Code are for personal use only and may not be reproduced without permission from the author of this poster.

Presented at the Society for Immunotherapy of Cancer (SITC) Annual Meeting, November 10-14, 2021.